Solving PK Challenges- Inhaled Peptide-Drug Conjugates allow for tunable PK profiles, maximizing efficacy whilst reducing dose and dosing frequency

Frank Guarnieri, Mariah Stewart, Dani M Stoltzfus, Eric Springman

Paint Therapeutics

T0930-08-49

Peptide-Drug Conjugate Solves PK Challenge for Potent Drug Candidate

PURPOSE

Improve PK properties of potent drug candidates

The desire to improve plasma half-life is two-fold, longer half-live equates to less frequent dosing for patients as well as improved efficacy and side-effect profiles.

OBJECTIVE

Demonstrate inhaled delivery of Peptide-Drug Conjugates improves PK through improved lung retention

The lung is an ideal delivery route due to its drug absorption features, but retaining drugs within the lungs has proven challenging.

METHODS

Peptide-drug conjugates

A series of peptide-drug conjugates consisting of an 11-amino acid GLP-1 receptor agonist (PT-5000) were made and tested. Exemplar data for PT-5005 is shown.

PK Study

Free drug PT-5000 and peptide-drug conjugate PT-5005 were administered oropharyngeally in mice and plasma levels of PT-5000 were measured at 0, 1, 2, 4, 8 and 24h post-dose.

Biodistribution Study

Cy5.5 labelled peptide-drug conjugate PT-5005-Cy5.5 was administered oropharyngeally to mice and bioluminescence in the chest cavity was measured at 0, 0.5, 1, 2, 4, 8, 24, 48, 72, 96, 120, 144, 168, 192, 216 & 240h post-dose.

Efficacy Study

Free drug PT-5000 and peptide-drug conjugate PT-5005 were administered oropharyngeally in diet induced obesity mice QOD 20 ug/mouse. Semaglutide was administered sc QD 0.016 mg/kg. Change in body weight was measured on Day 27 of the study.

RESULTS

PK Study

- PT-5005 concentration 8 hours post-dose was equivalent to Cmax (8 ng/mL).
- PT-5000 exhibited standard exponentially decay and was undetectable 8 hours post-dose.

Biodistribution Study

- PT-5005 – Cy5.5 PT-5000 exhibited a very long lung residence time with a calculated elimination half-life of 1.7d (not shown) compared to 1.1d for PT-5000.

Efficacy Study

- PT-5005 achieved a body weight change equivalent to semaglutide on Day 27 of the study.
- PT-5000 did not achieve a body weight change and was equivalent to vehicle.

CONCLUSIONS

Peptide-drug conjugate solves PK challenge of potent drug candidate

- Modulation of the cleavable linker in peptide-drug conjugates can tune PK profile of drug candidates
- Biodistribution study confirms that peptide-drug conjugate have long lung residence time
- Formation of peptide-drug conjugate PT-5005 enables PT-5000 to be efficacious in a diet-induced obesity mouse model, with efficacy equivalent to the positive control semaglutide.
- PT-5005 was able to be administered less frequently than semaglutide, indicating that controlling the release of the drug from the Paint peptide impacts the concentration of drug in systemic circulation over time.

PARTNER WITH US

Contact Ron Sarkar at rsarkar@painttherapeutics.com to start the conversation, or visit us at www.painttherapeutics.com

paint therapeutics

aaps PharmSci 360

PEPTIDE-DRUG CONJUGATE RELEASES DRUG FROM LUNGS INTO SYSTEMIC CIRCULATION

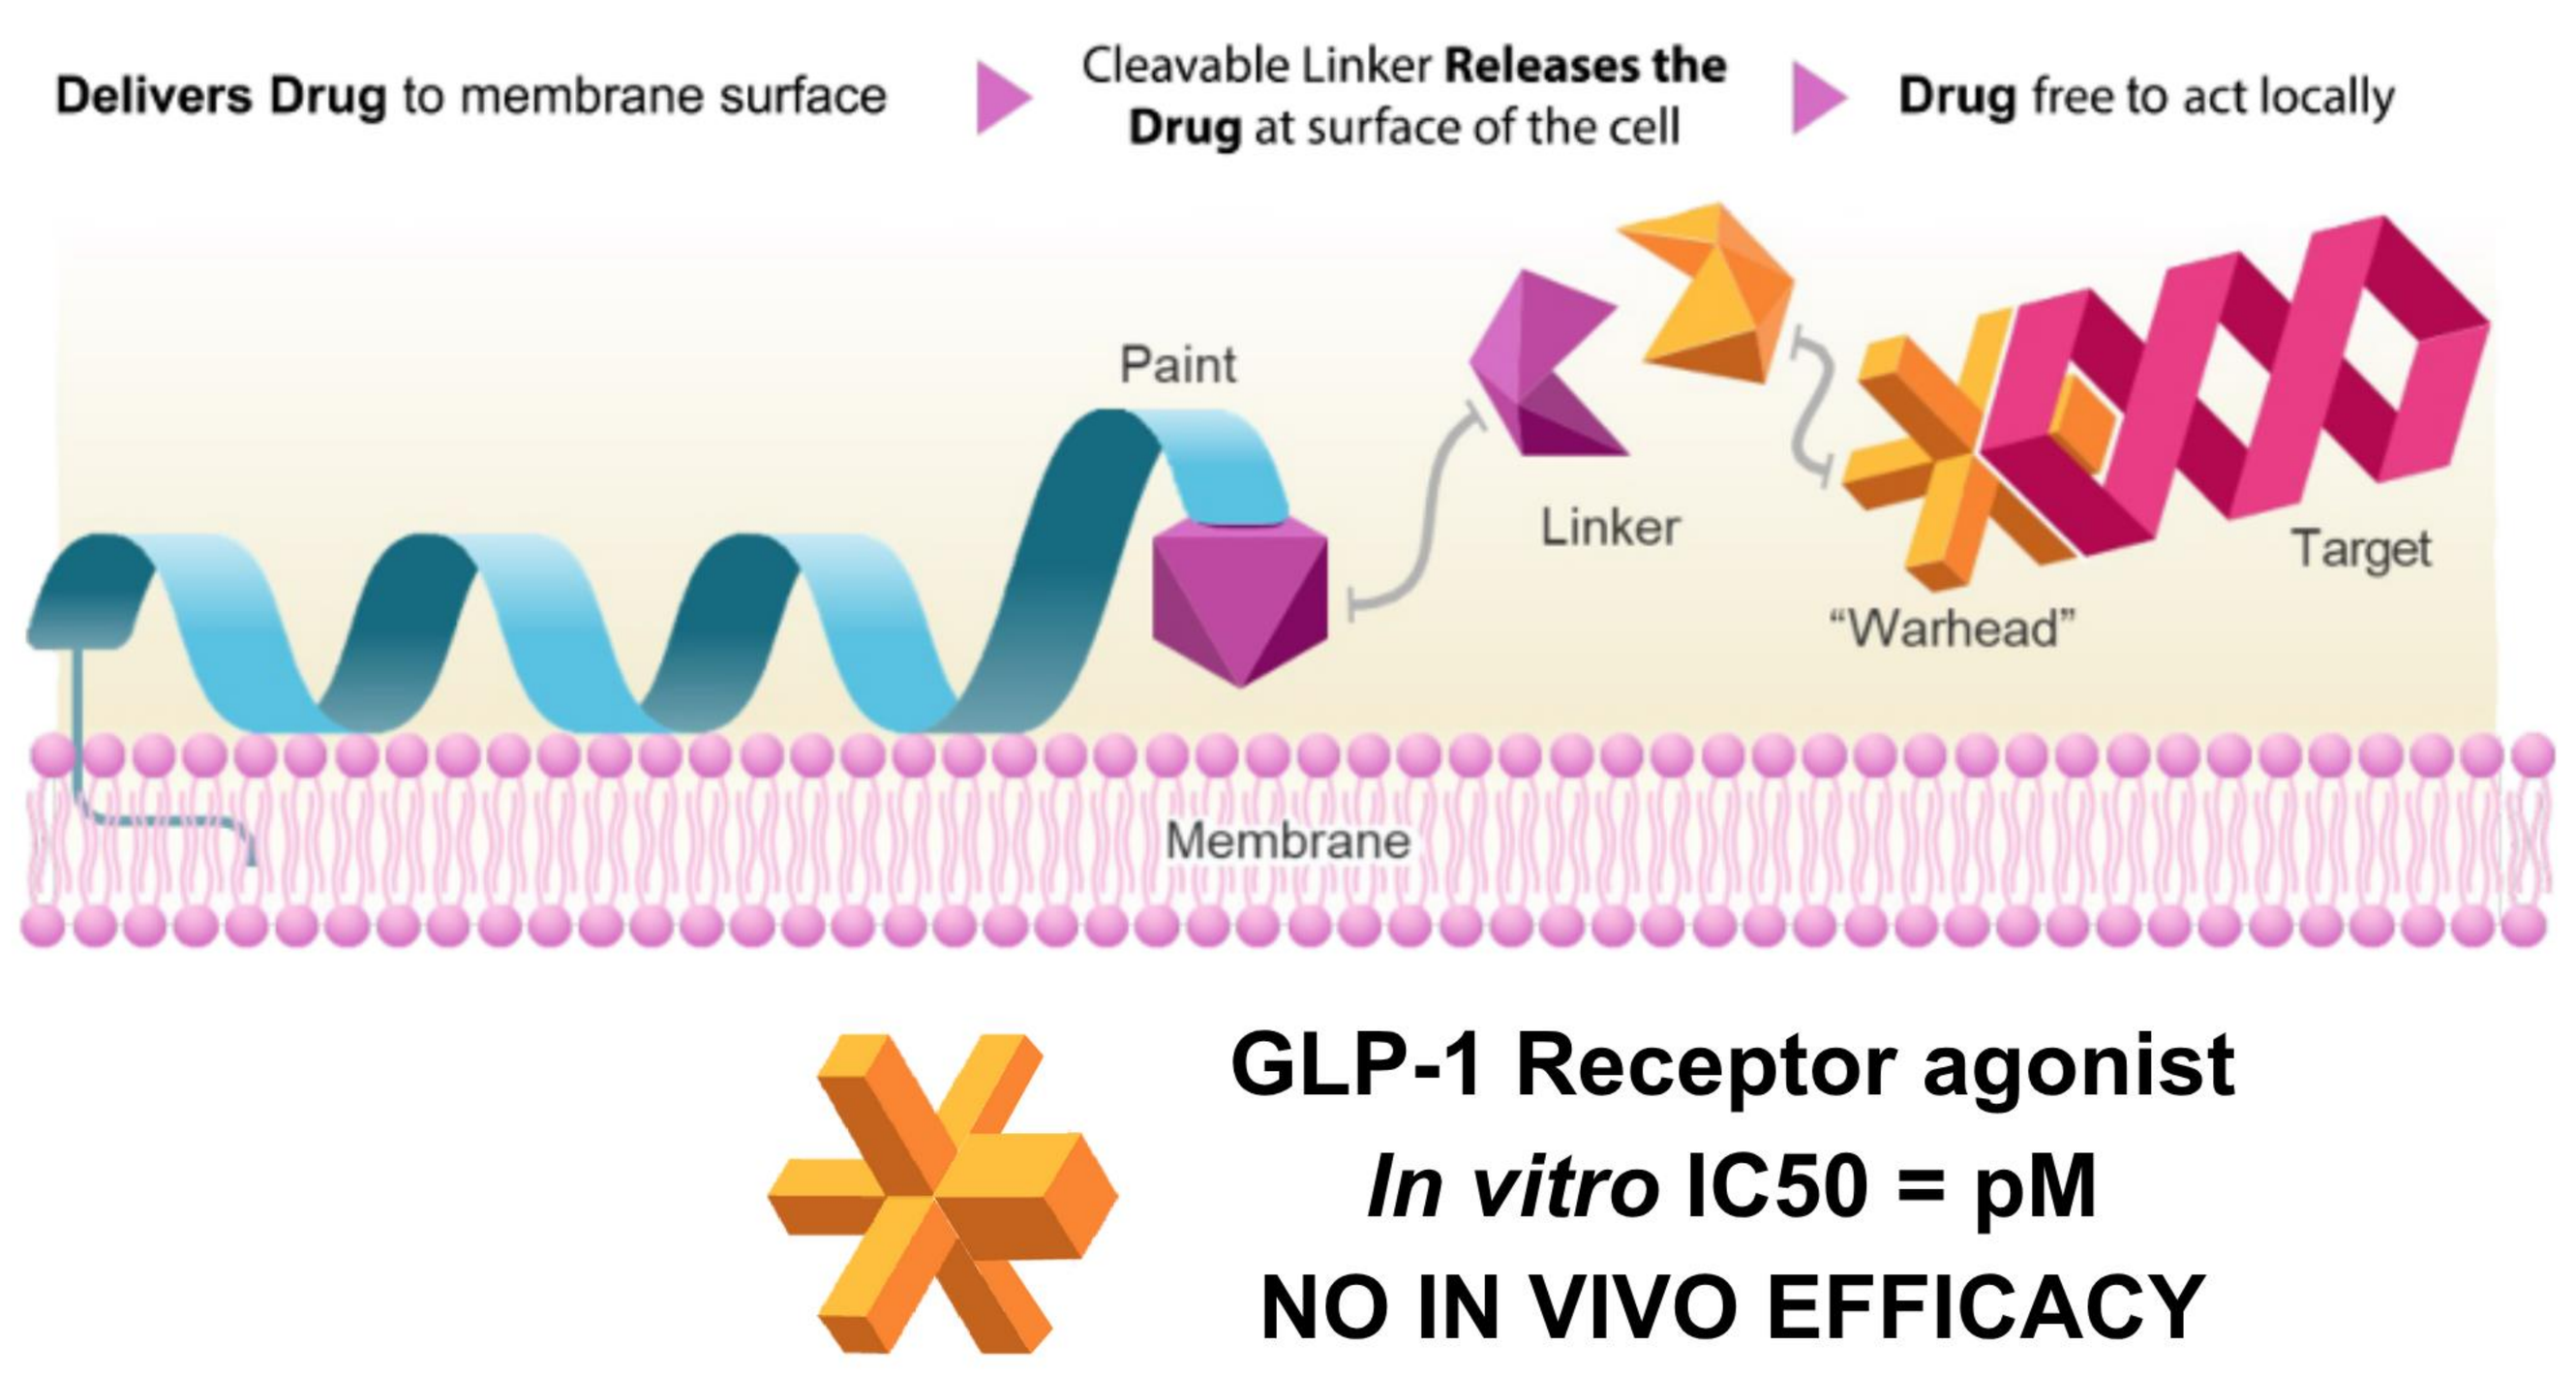
Peptide Drug Conjugate Mechanism of Action
Delivers Drug to membrane surface
Cleavable Linker Releases the Drug at surface of the cell
Drug free to act locally
Paint
Linker
"Warhead"
Target
Membrane
GLP-1 Receptor agonist
In vitro IC50 = pM
NO IN VIVO EFFICACY

PEPTIDE-DRUG CONJUGATE IMPROVES PK OF DRUG CANDIDATE

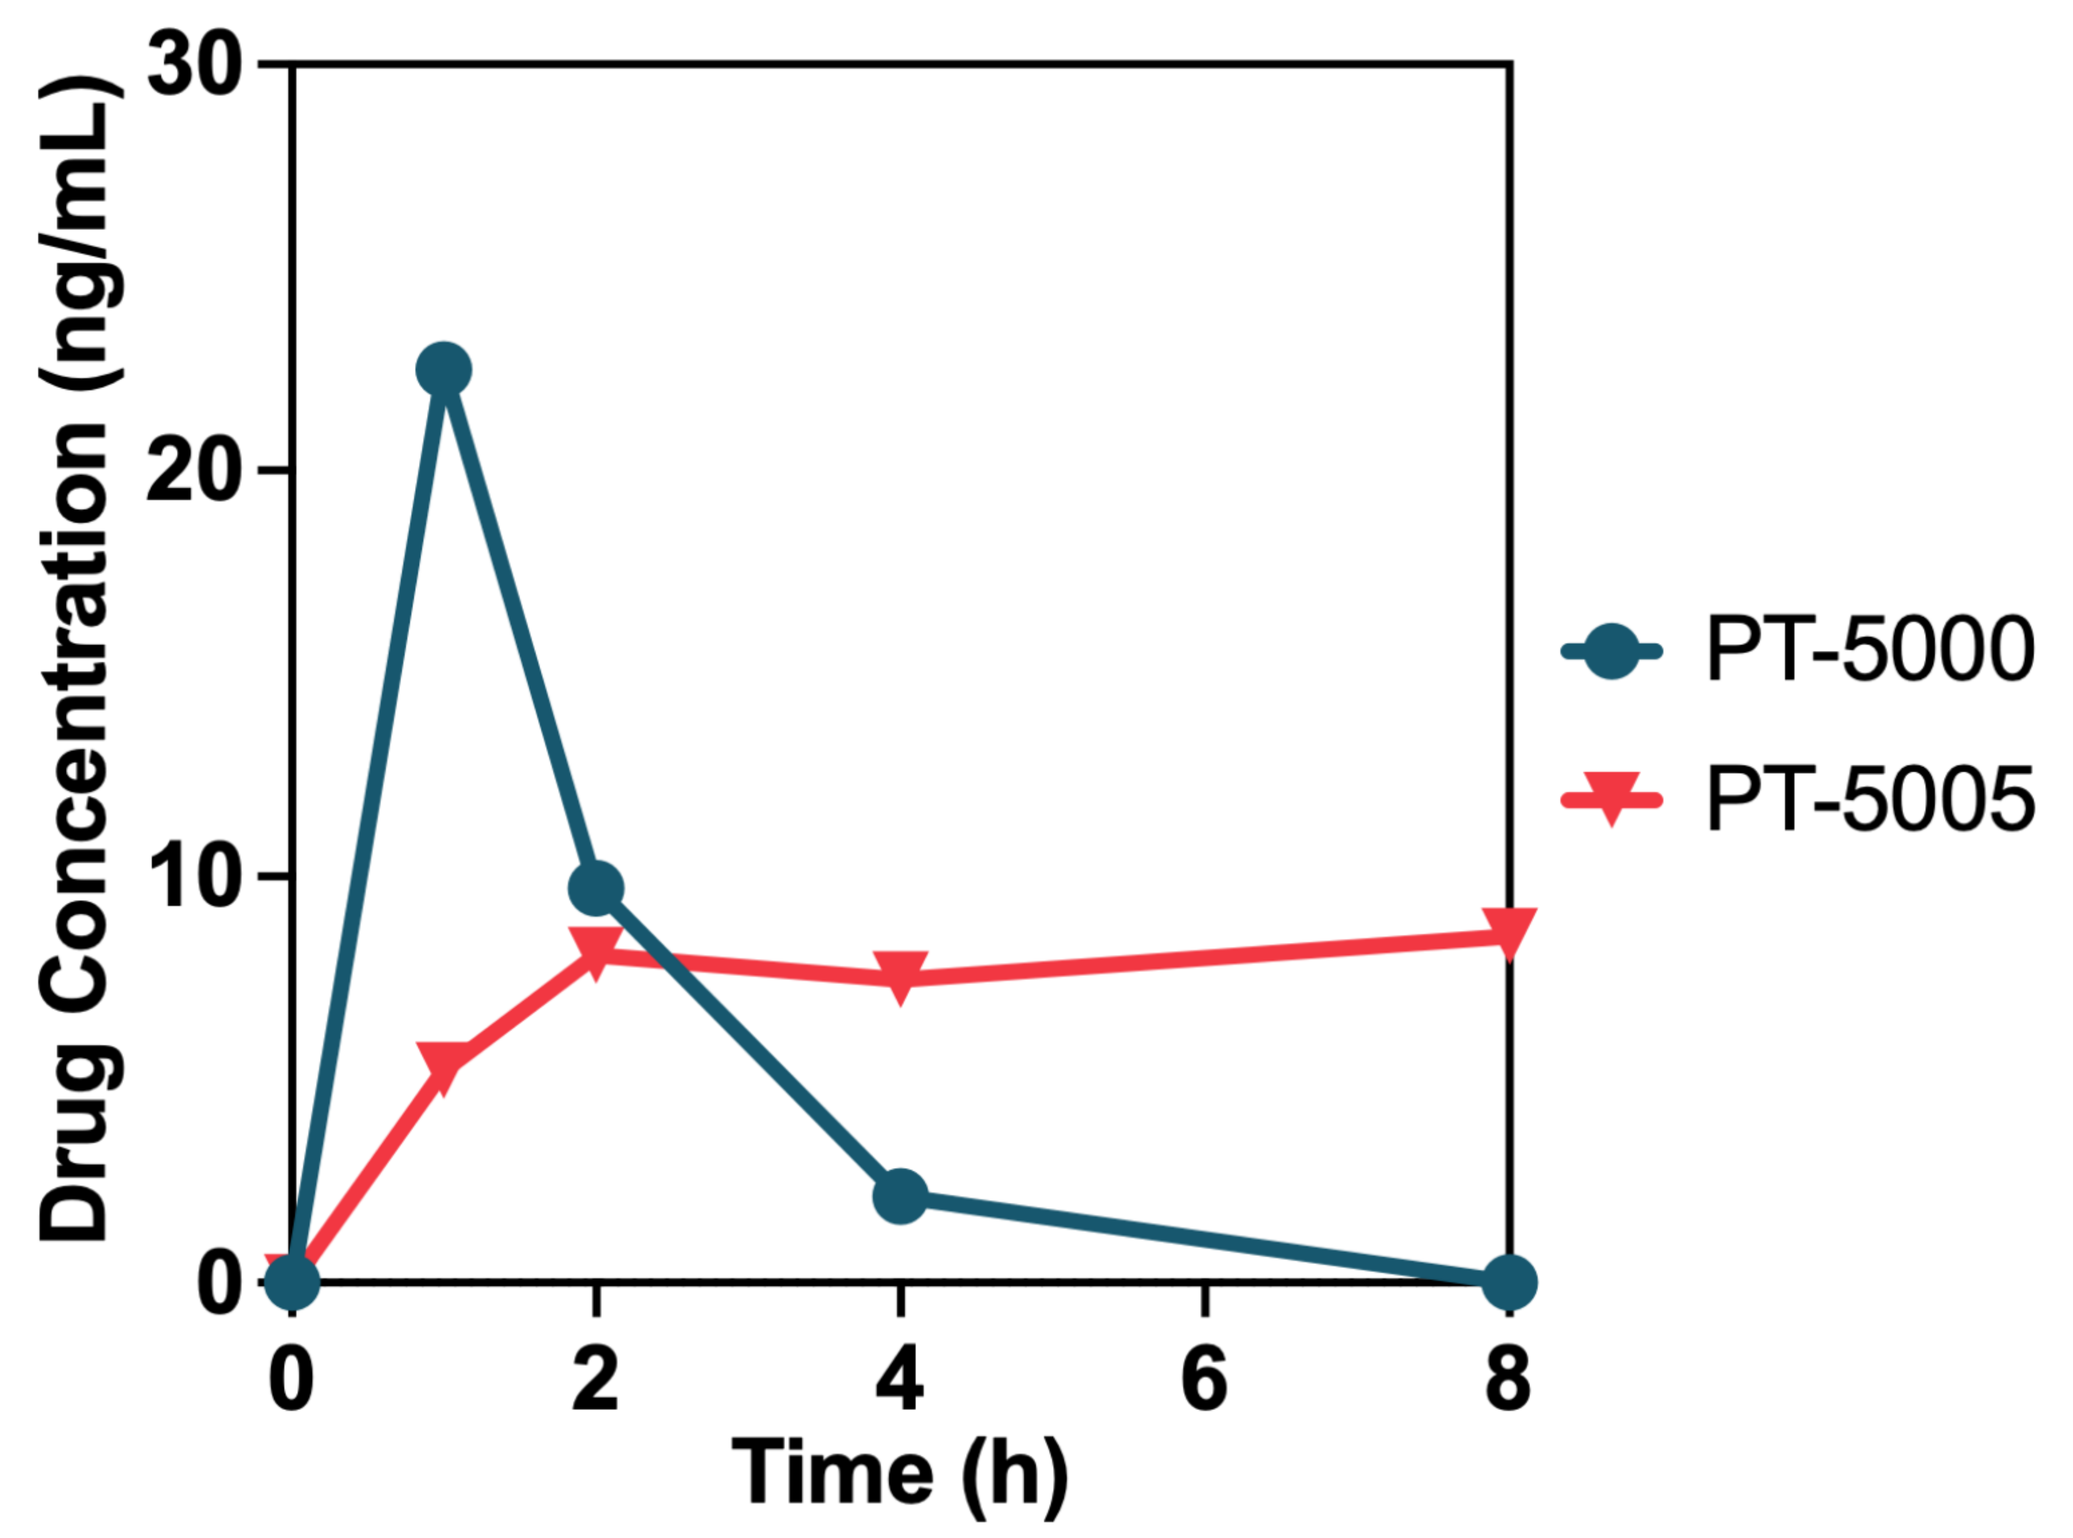
Concentration of Drug in Plasma
Drug Concentration (ng/mL)
30
20
10
0
0
2
4
6
8
Time (h)
PT-5000
PT-5005

PEPTIDE-DRUG CONJUGATE HAS VERY LONG LUNG RESIDENCE TIME

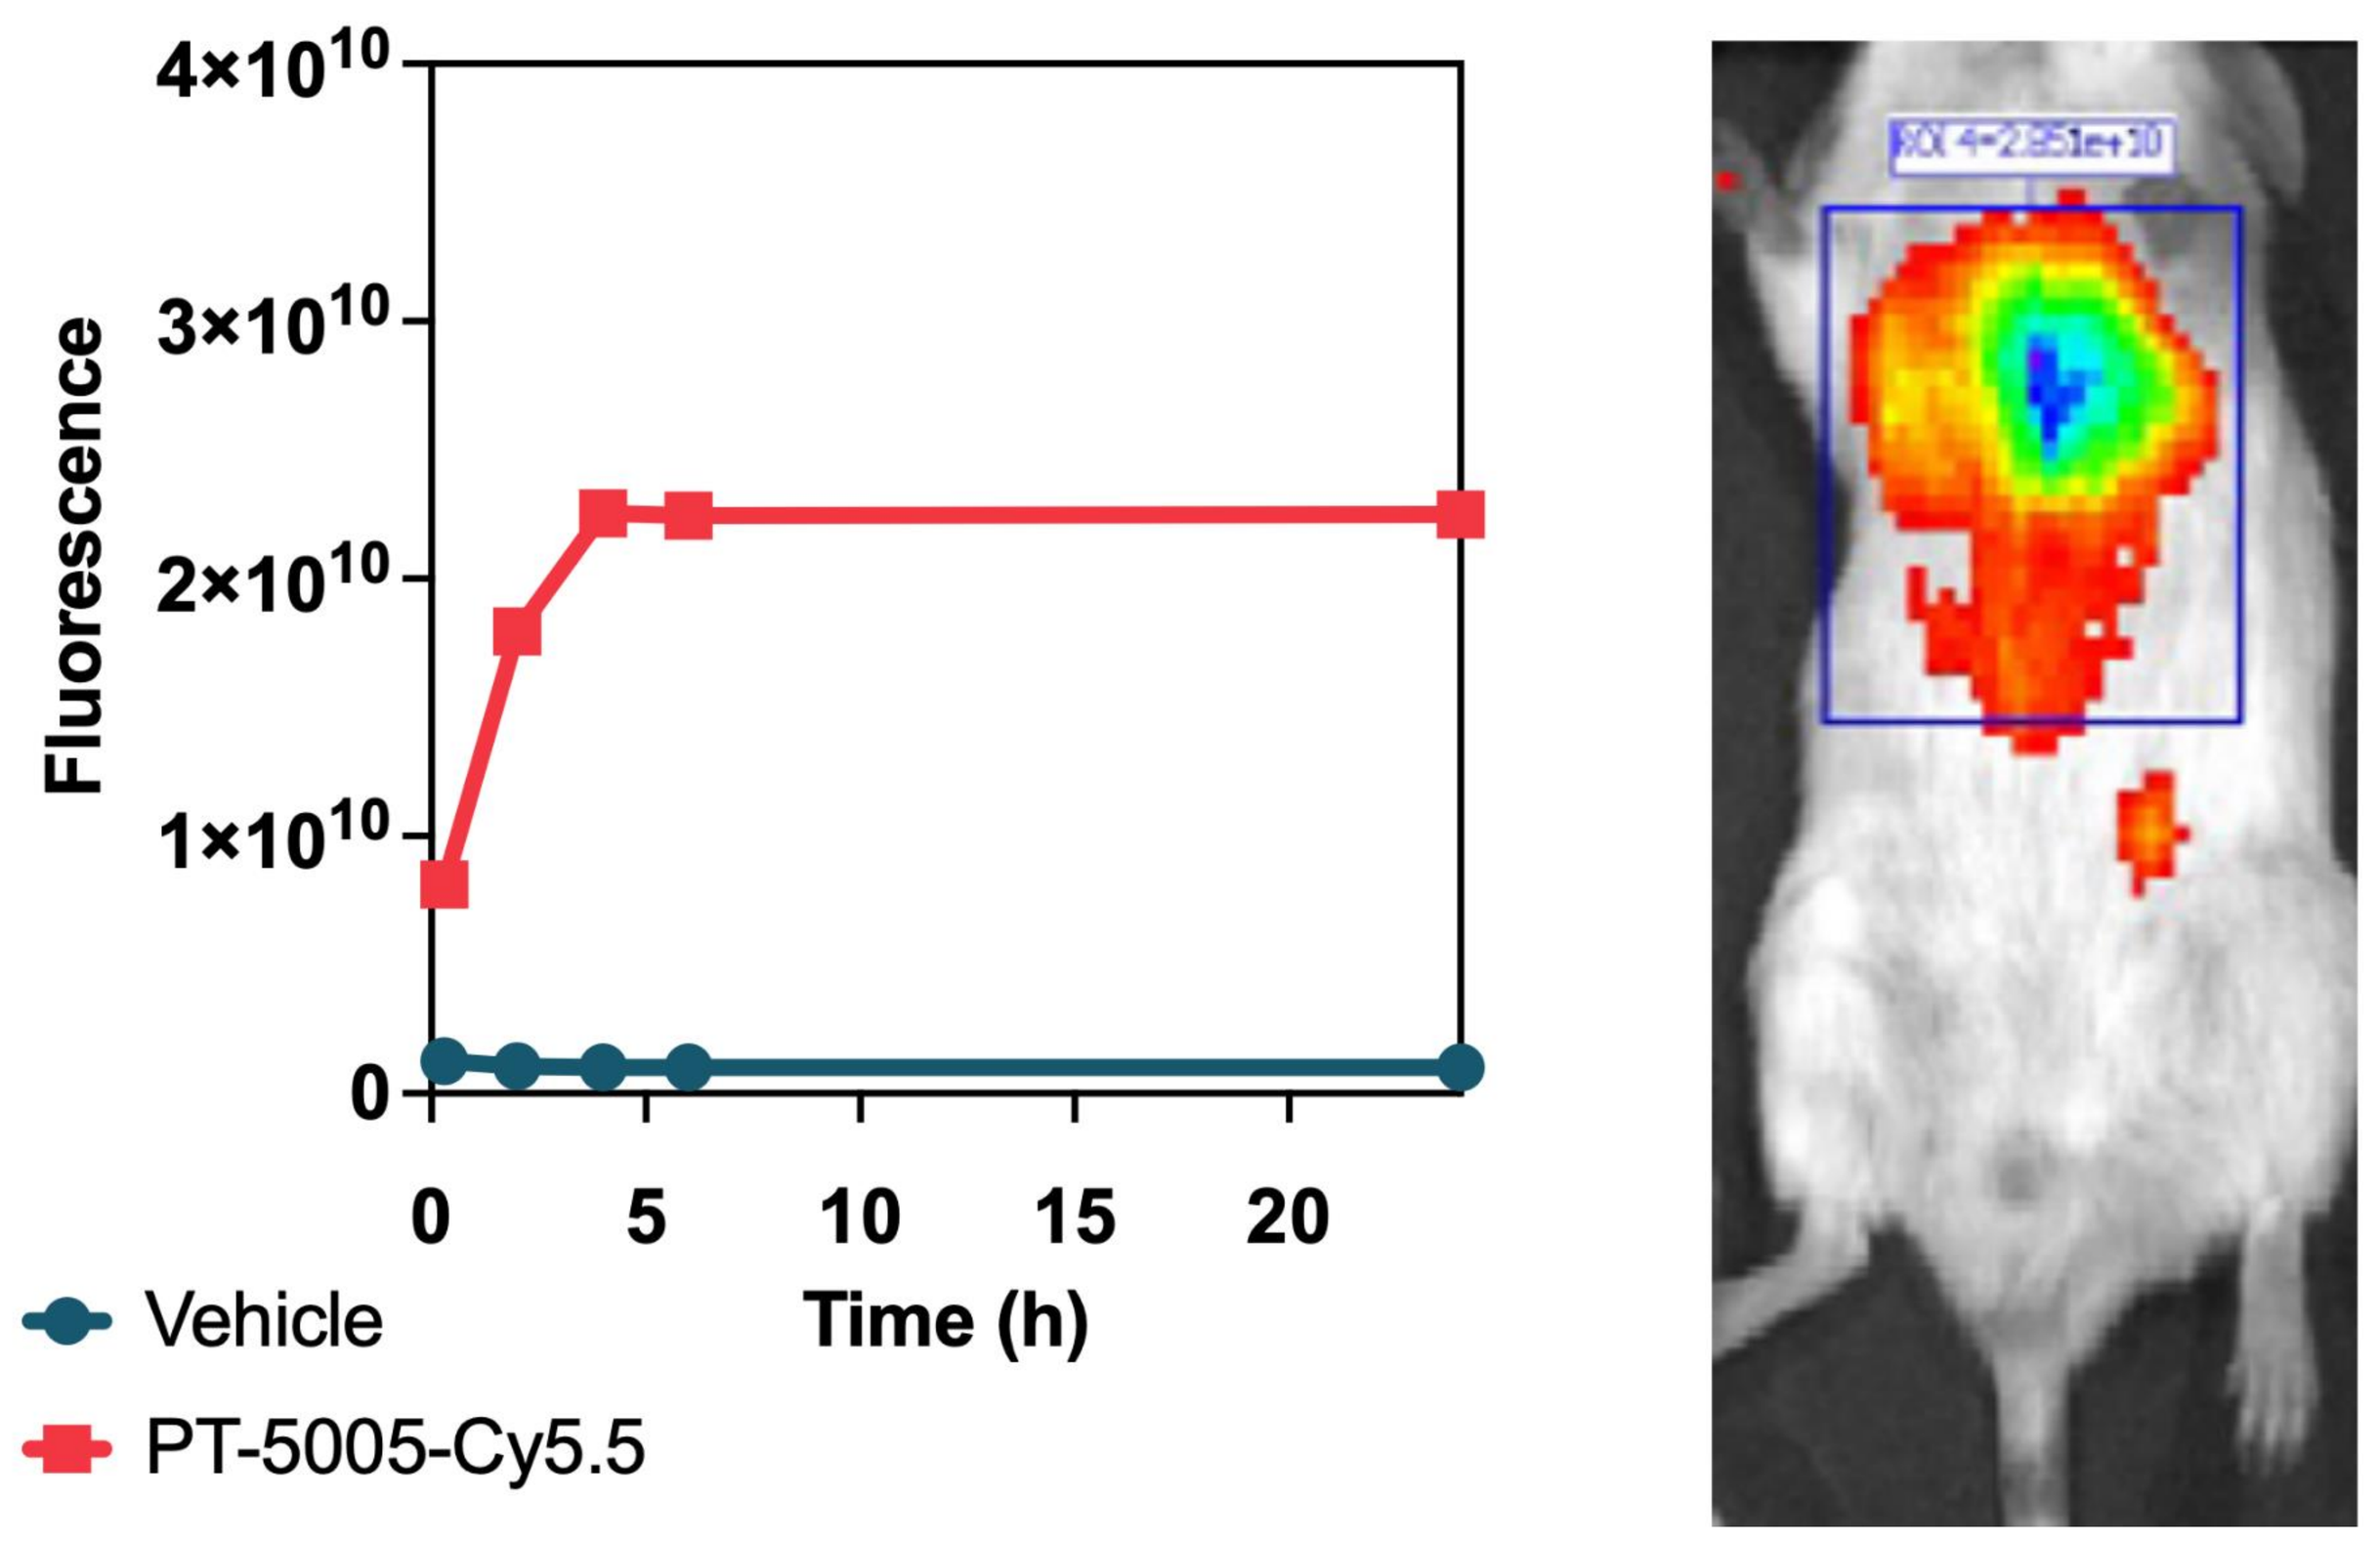
Fluorescence Intensity in Lungs
Fluorescence
4×10^10
3×10^10
2×10^10
1×10^10
0
0
5
10
15
20
Time (h)
Vehicle
PT-5005-Cy5.5

PEPTIDE-DRUG CONJUGATE ACHIEVES EFFICACY EQUIVALENT TO BLOCKBUSTER DRUG

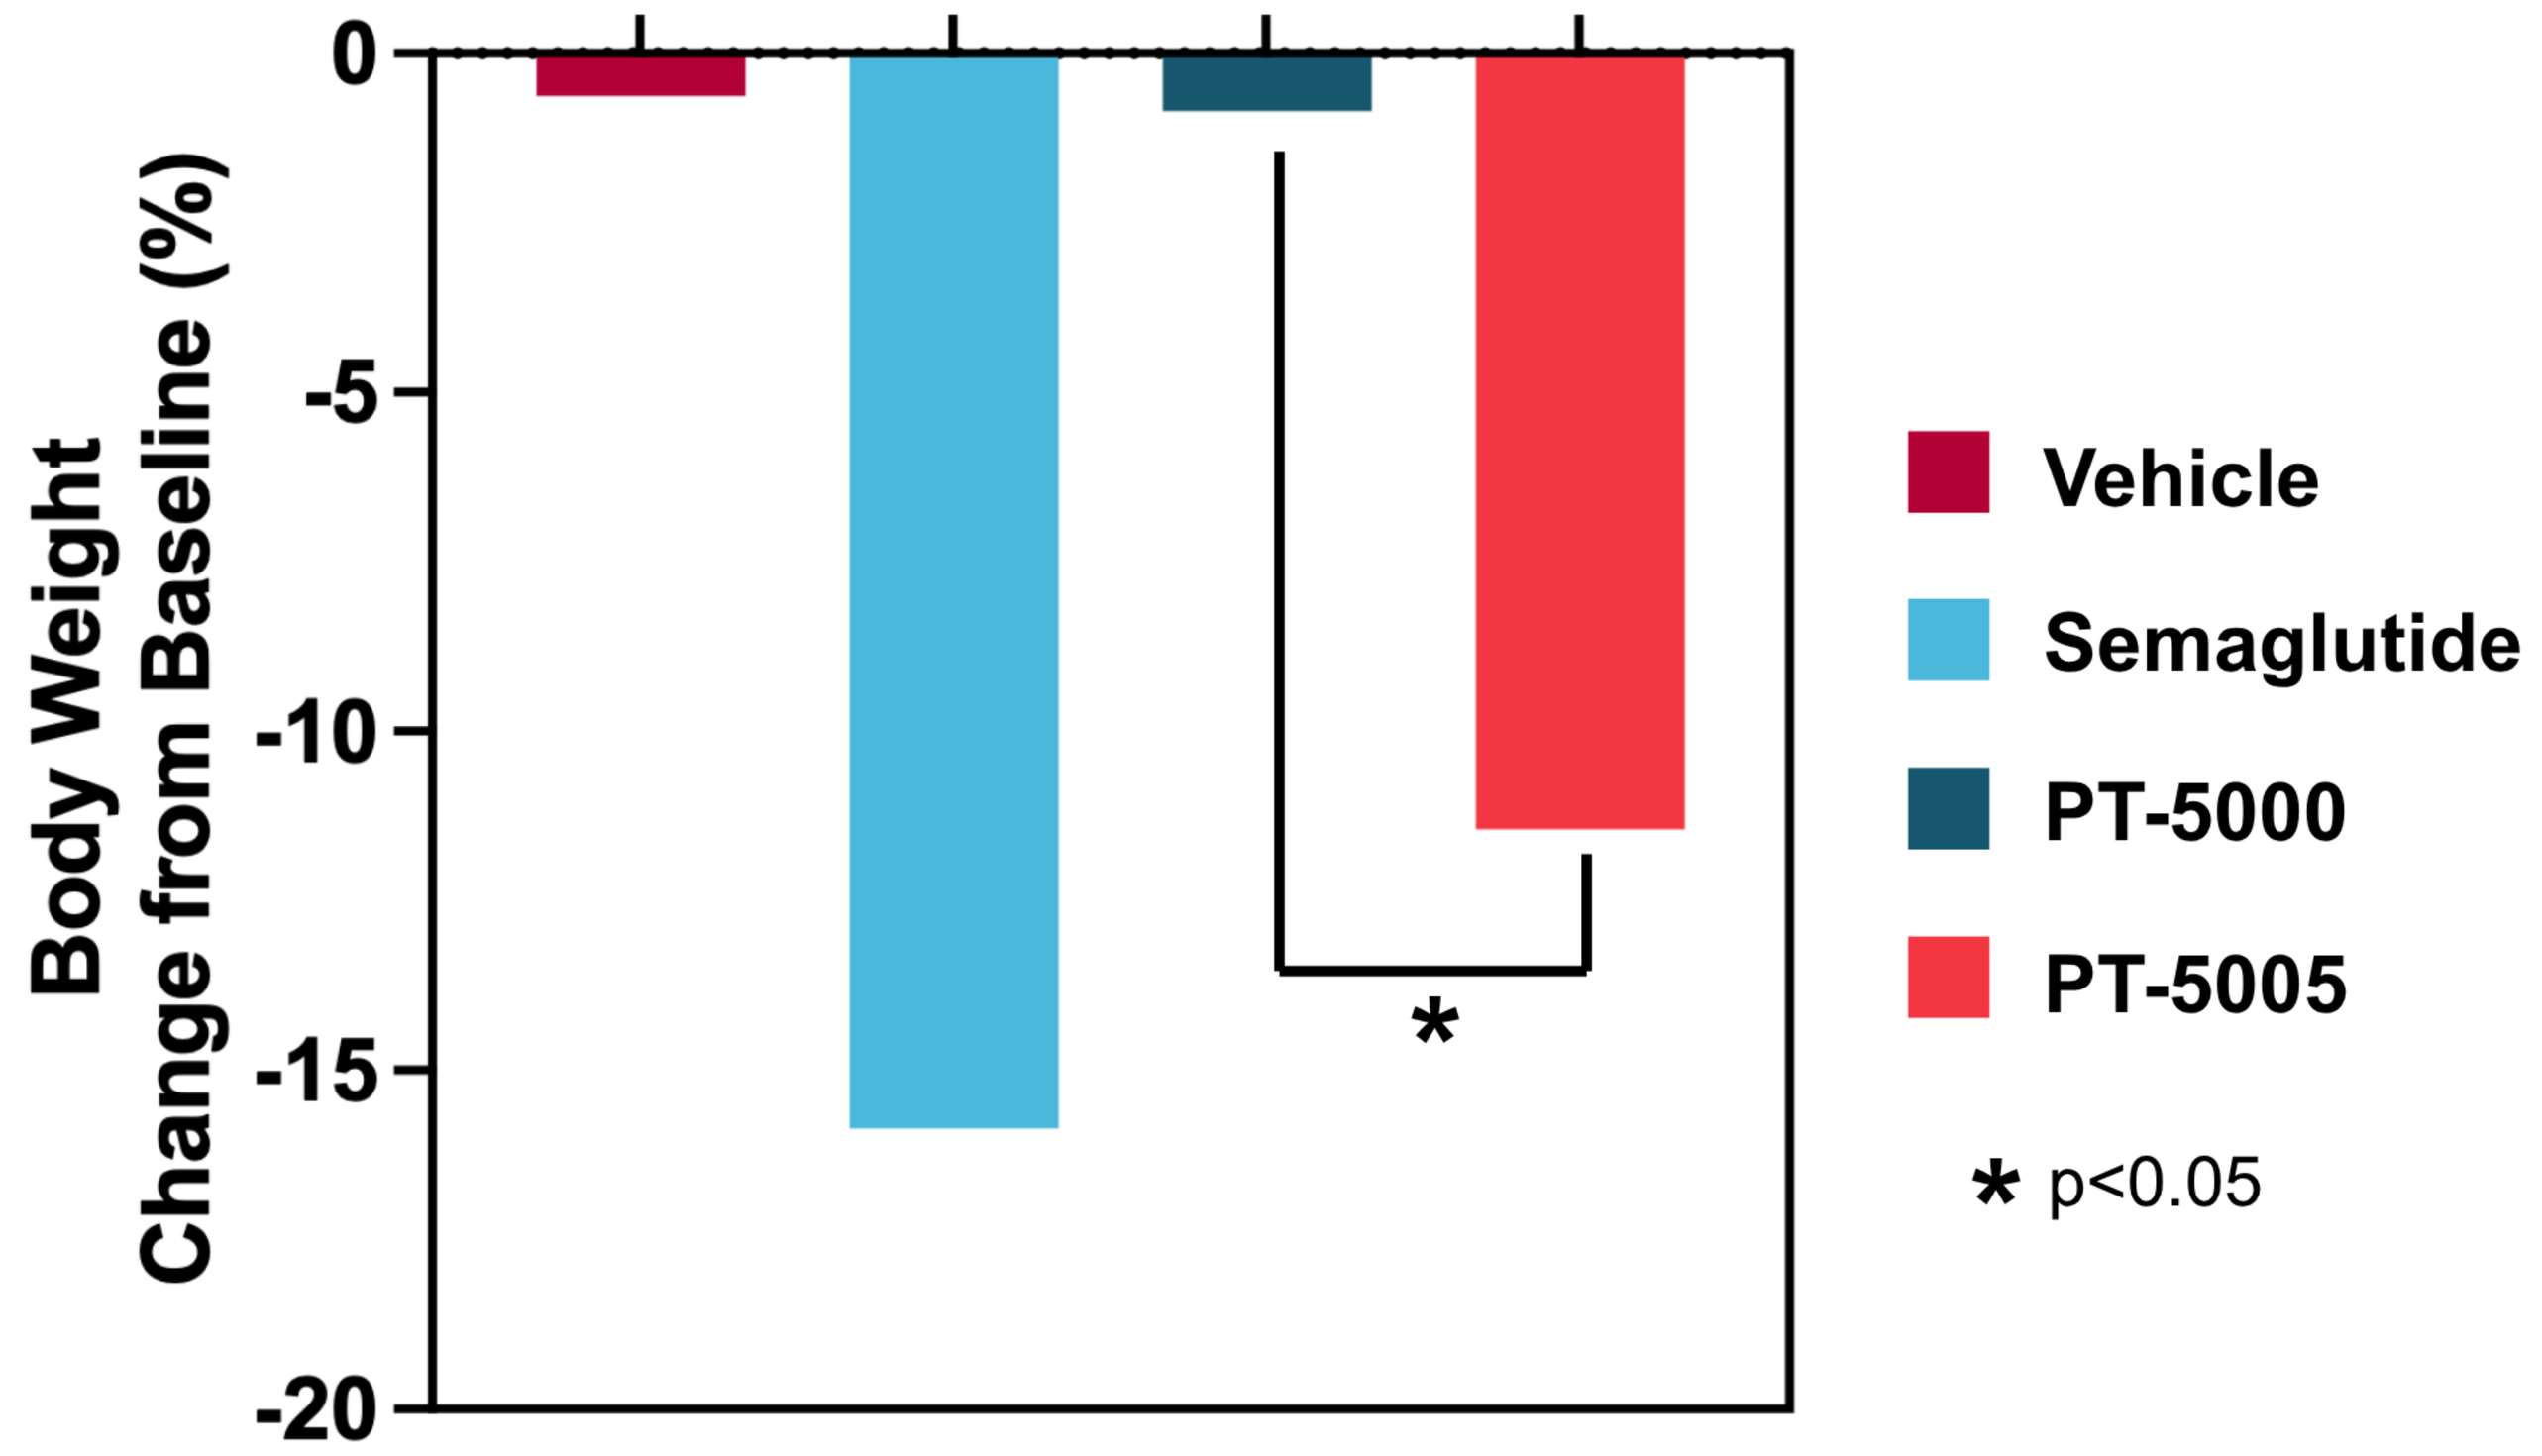
Body Weight Change Day 27
Body Weight Change from Baseline (%)
0
-5
-10
-15
-20
*
Vehicle
Semaglutide
PT-5000
PT-5005
* p<0.05

CONTACT INFORMATION: Eric Springman, espringman@painttherapeutics.com